125P

MSS/KRAS^{G12X} CRC Depends on CBM Signalosome for Survival - A Discovery that May Transform KRAS^{G12X} CRC Therapy

Fang Wang[1], Yi Yao[1], Kevin Ling[1], Huili Xia[1], Yilin Qi[1], Huadong Sun[1], Sharon Wu[2], Wei Liu[1], Gerry Harriman[1]

HotSpot Therapeutics, Inc, One Design Center Place, Suite 19-600, Boston, MA 02210, USA; . Caris Life Science, 750 W John Carpenter Fwy, Suite 800, Irving, TX 75039, USA

Introduction

KRAS activation is a prominent genetic feature of colorectal cancer (CRC), however, CRC patients did not demonstrate deep or durable responses when treated with KRAS inhibitors.

We and others have observed that KRAS inhibitors, while potently inhibiting cancer proliferation, do not induce strong apoptosis *in vitro*, *in vivo* or in patients with CRC. This has led to limited and non-durable clinical response of KRASi, because tumor cells remain alive and soon relapse or develop resistance.

We discovered that MSS/KRAS^{G12} CRC depends on CBM signalosome for survival. Inhibition of CBM selectively induces selective and potent apoptosis in KRAS^{G12} CRC.

CBM Signalosome, the Pro-Survival Hub for Cancer Progression

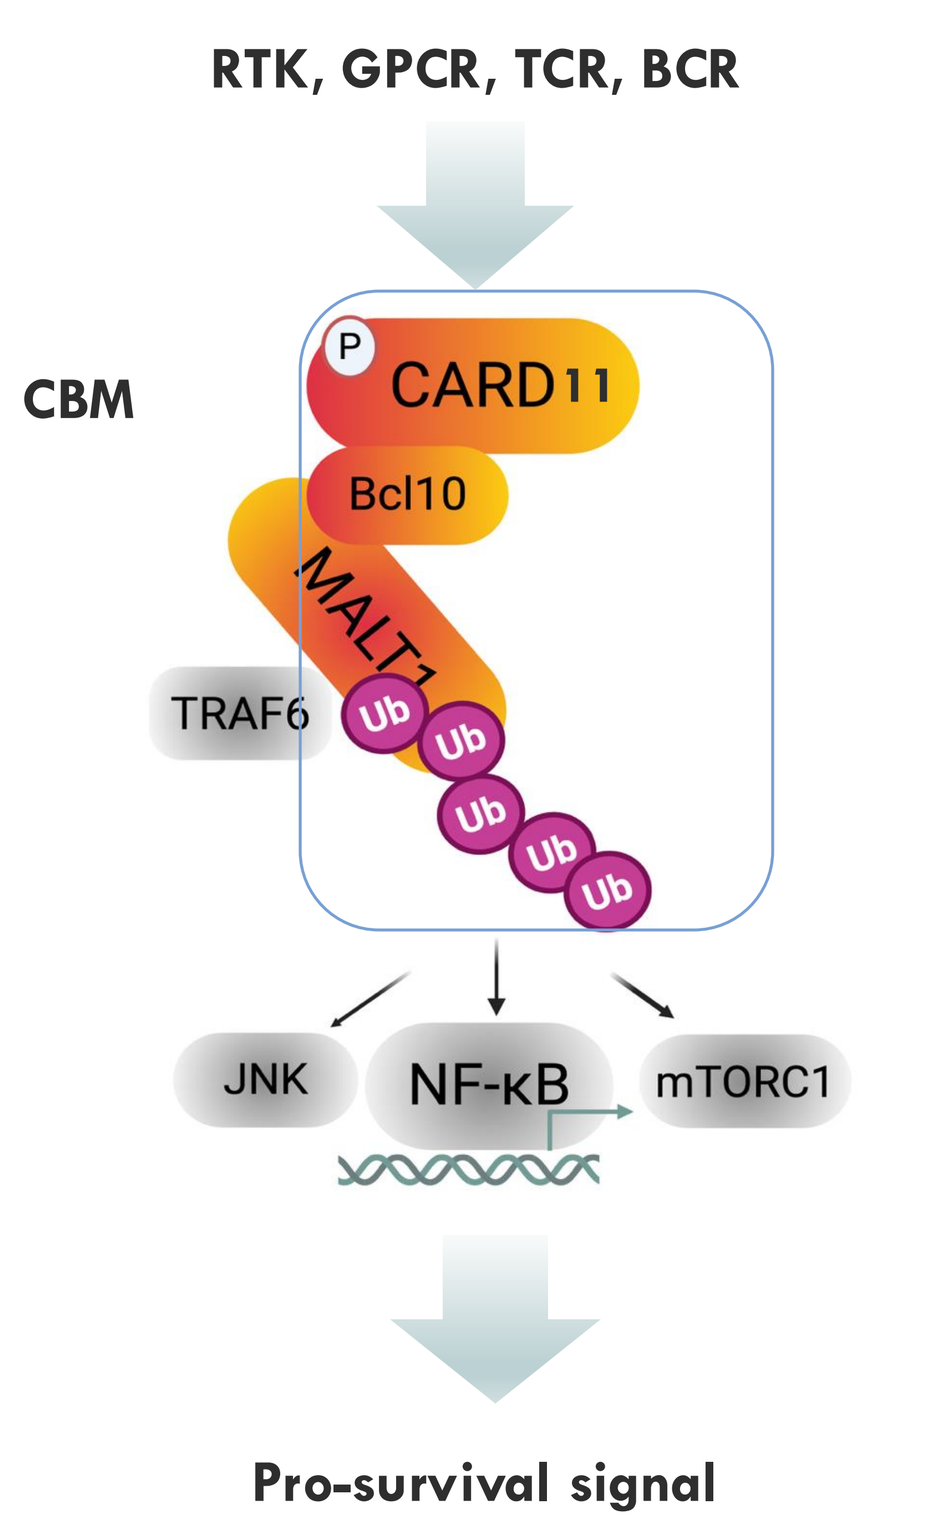
RTK, GPCR, TCR, BCR
CBM
P
CARD11
Bcl10
MALT1
TRAF6
Ub
JNK
NF-κB
mTORC1
Pro-survival signal

CBM signalosome: CARD11-BCL10-MALT1 complex.

CBM transduces upstream growth & survival signals via a series of phosphorylation and ubiquitination reactions, and activates multiple pro-survival pathways, including canonical NFκB, mTORC1 and JNK.

In cancer, upstream GoFs (BTK, PKC, etc), CARD11 GoF mutations, or CARD11 overexpression activate CBM signalosome, promoting cancer progression.

Lack of Potent Apoptosis Could Explain the Limited and Non-Durable Response of KRASi in CRC

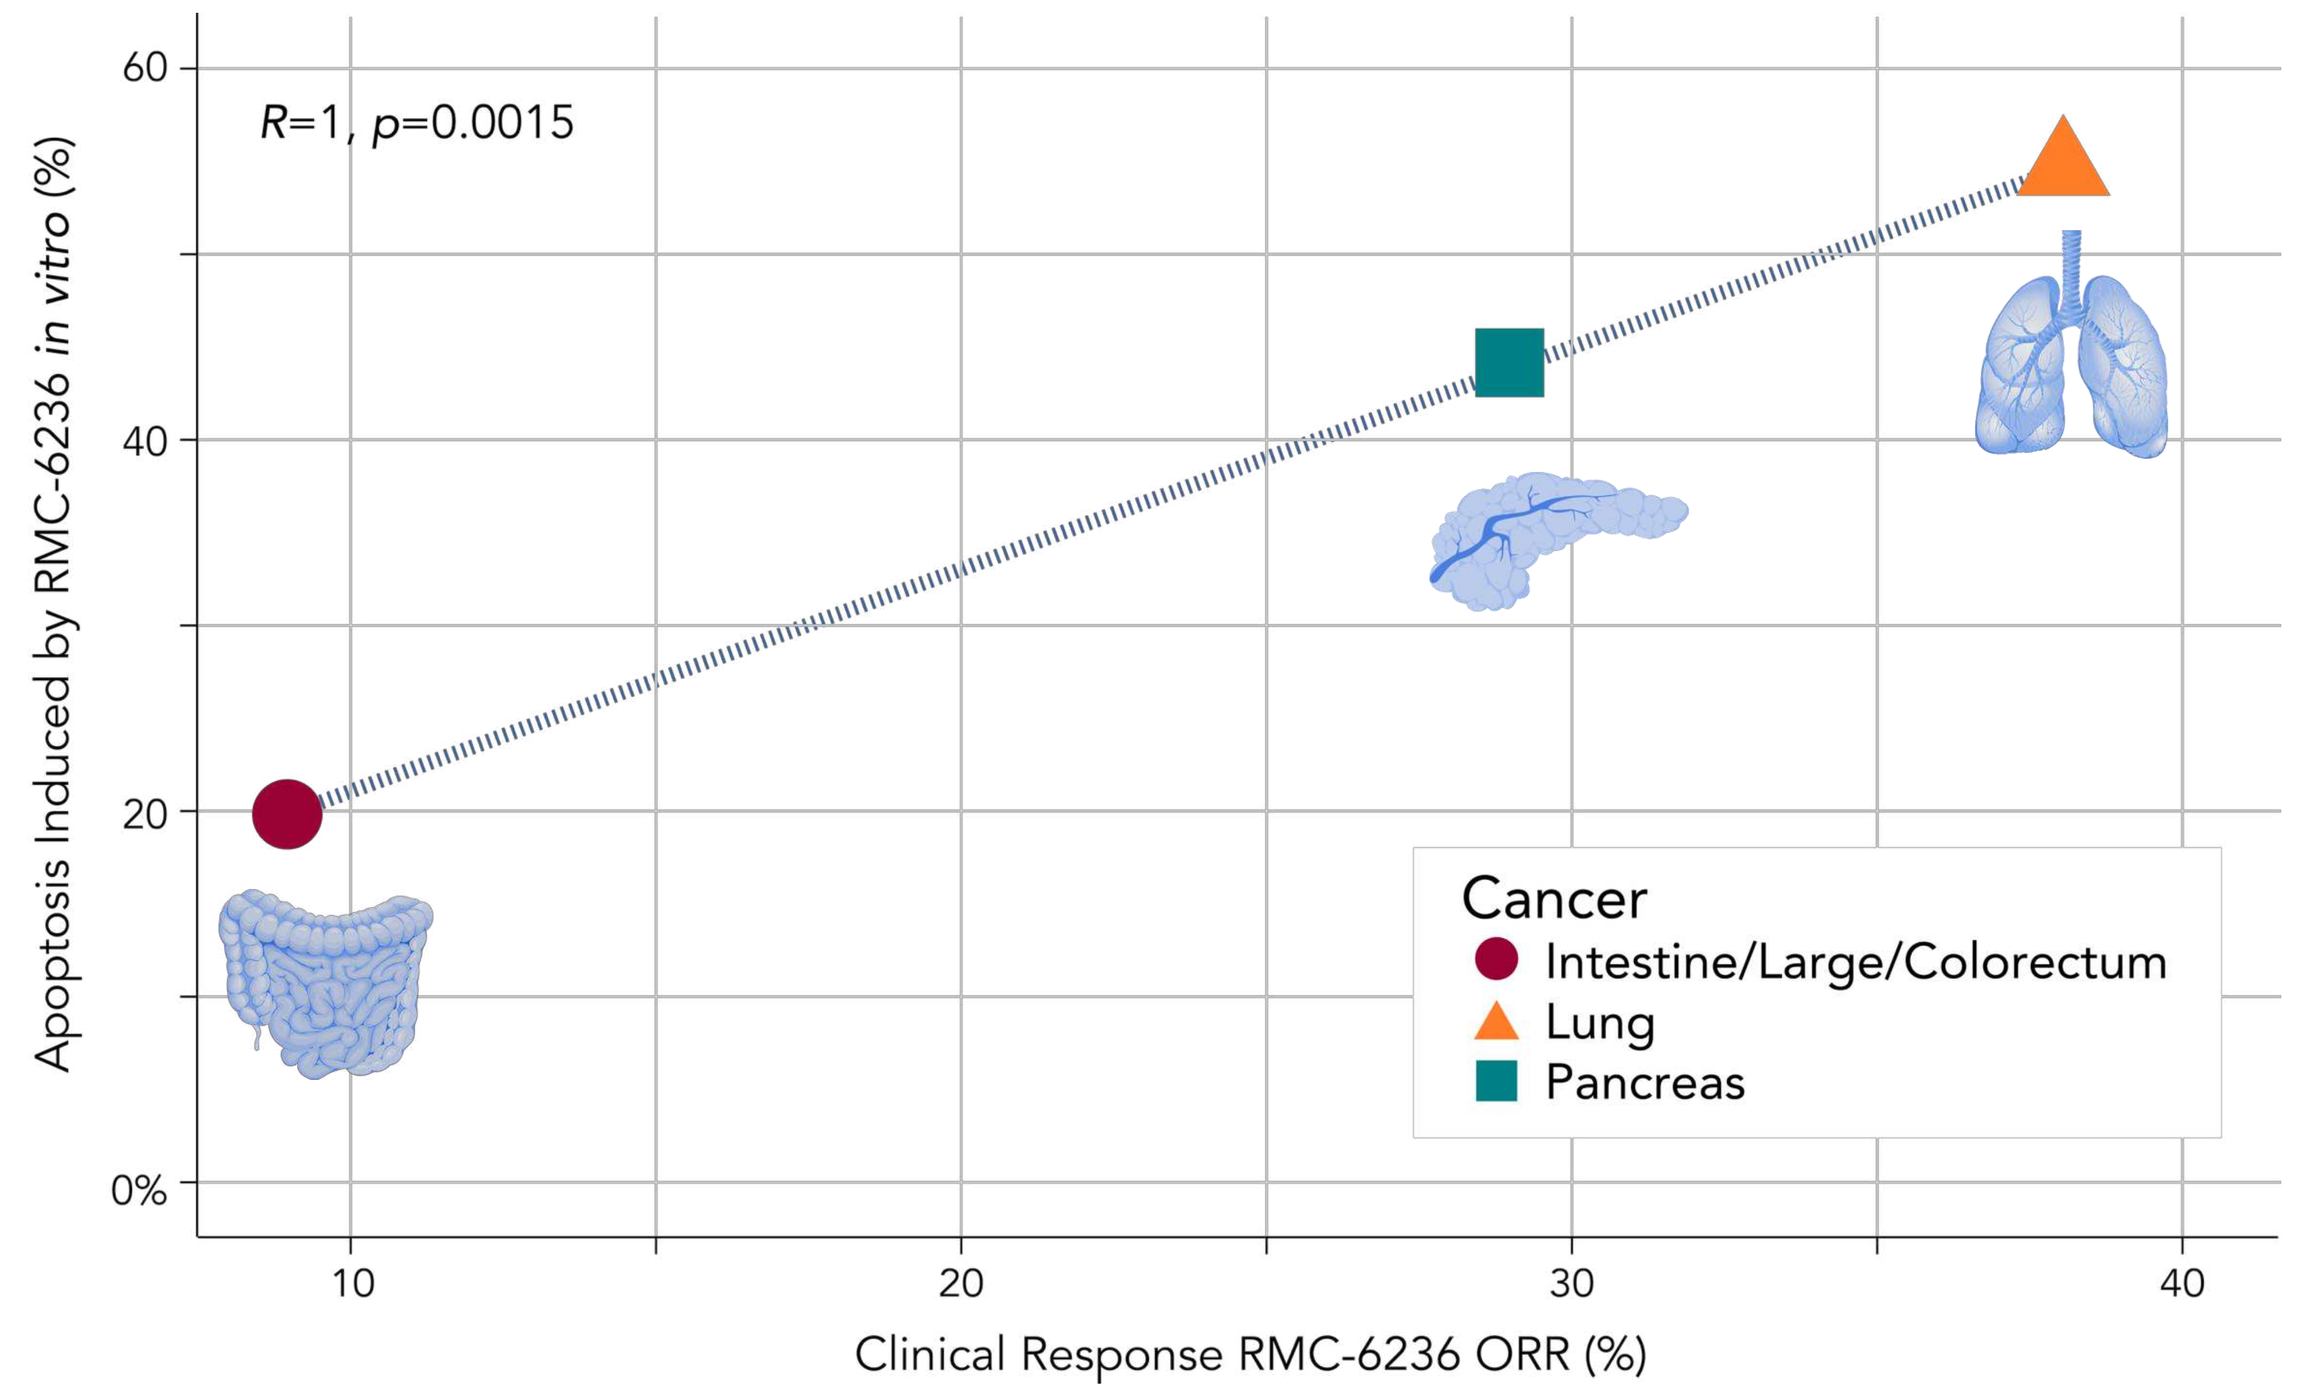
R=1, p=0.0015
Apoptosis Induced by RMC-6236 in vitro (%)
Clinical Response RMC-6236 ORR (%)
Cancer
Intestine/Large/Colorectum
Lung
Pancreas

In vitro data was from Revolution Medicine and HotSpot's data; the rate represents the percentage of cell lines responding to KRASi induced anti-proliferation or apoptosis in a cell panel screen.

Results

Figure 1. In CRC, High CARD11 CRC had Highest KRAS Activity and Shorter KRASi Time-on-Treatment

A Study of > 20,000 CRC Patients Database Revealed CARD11 CNA or Amp in >50% of MSS CRC

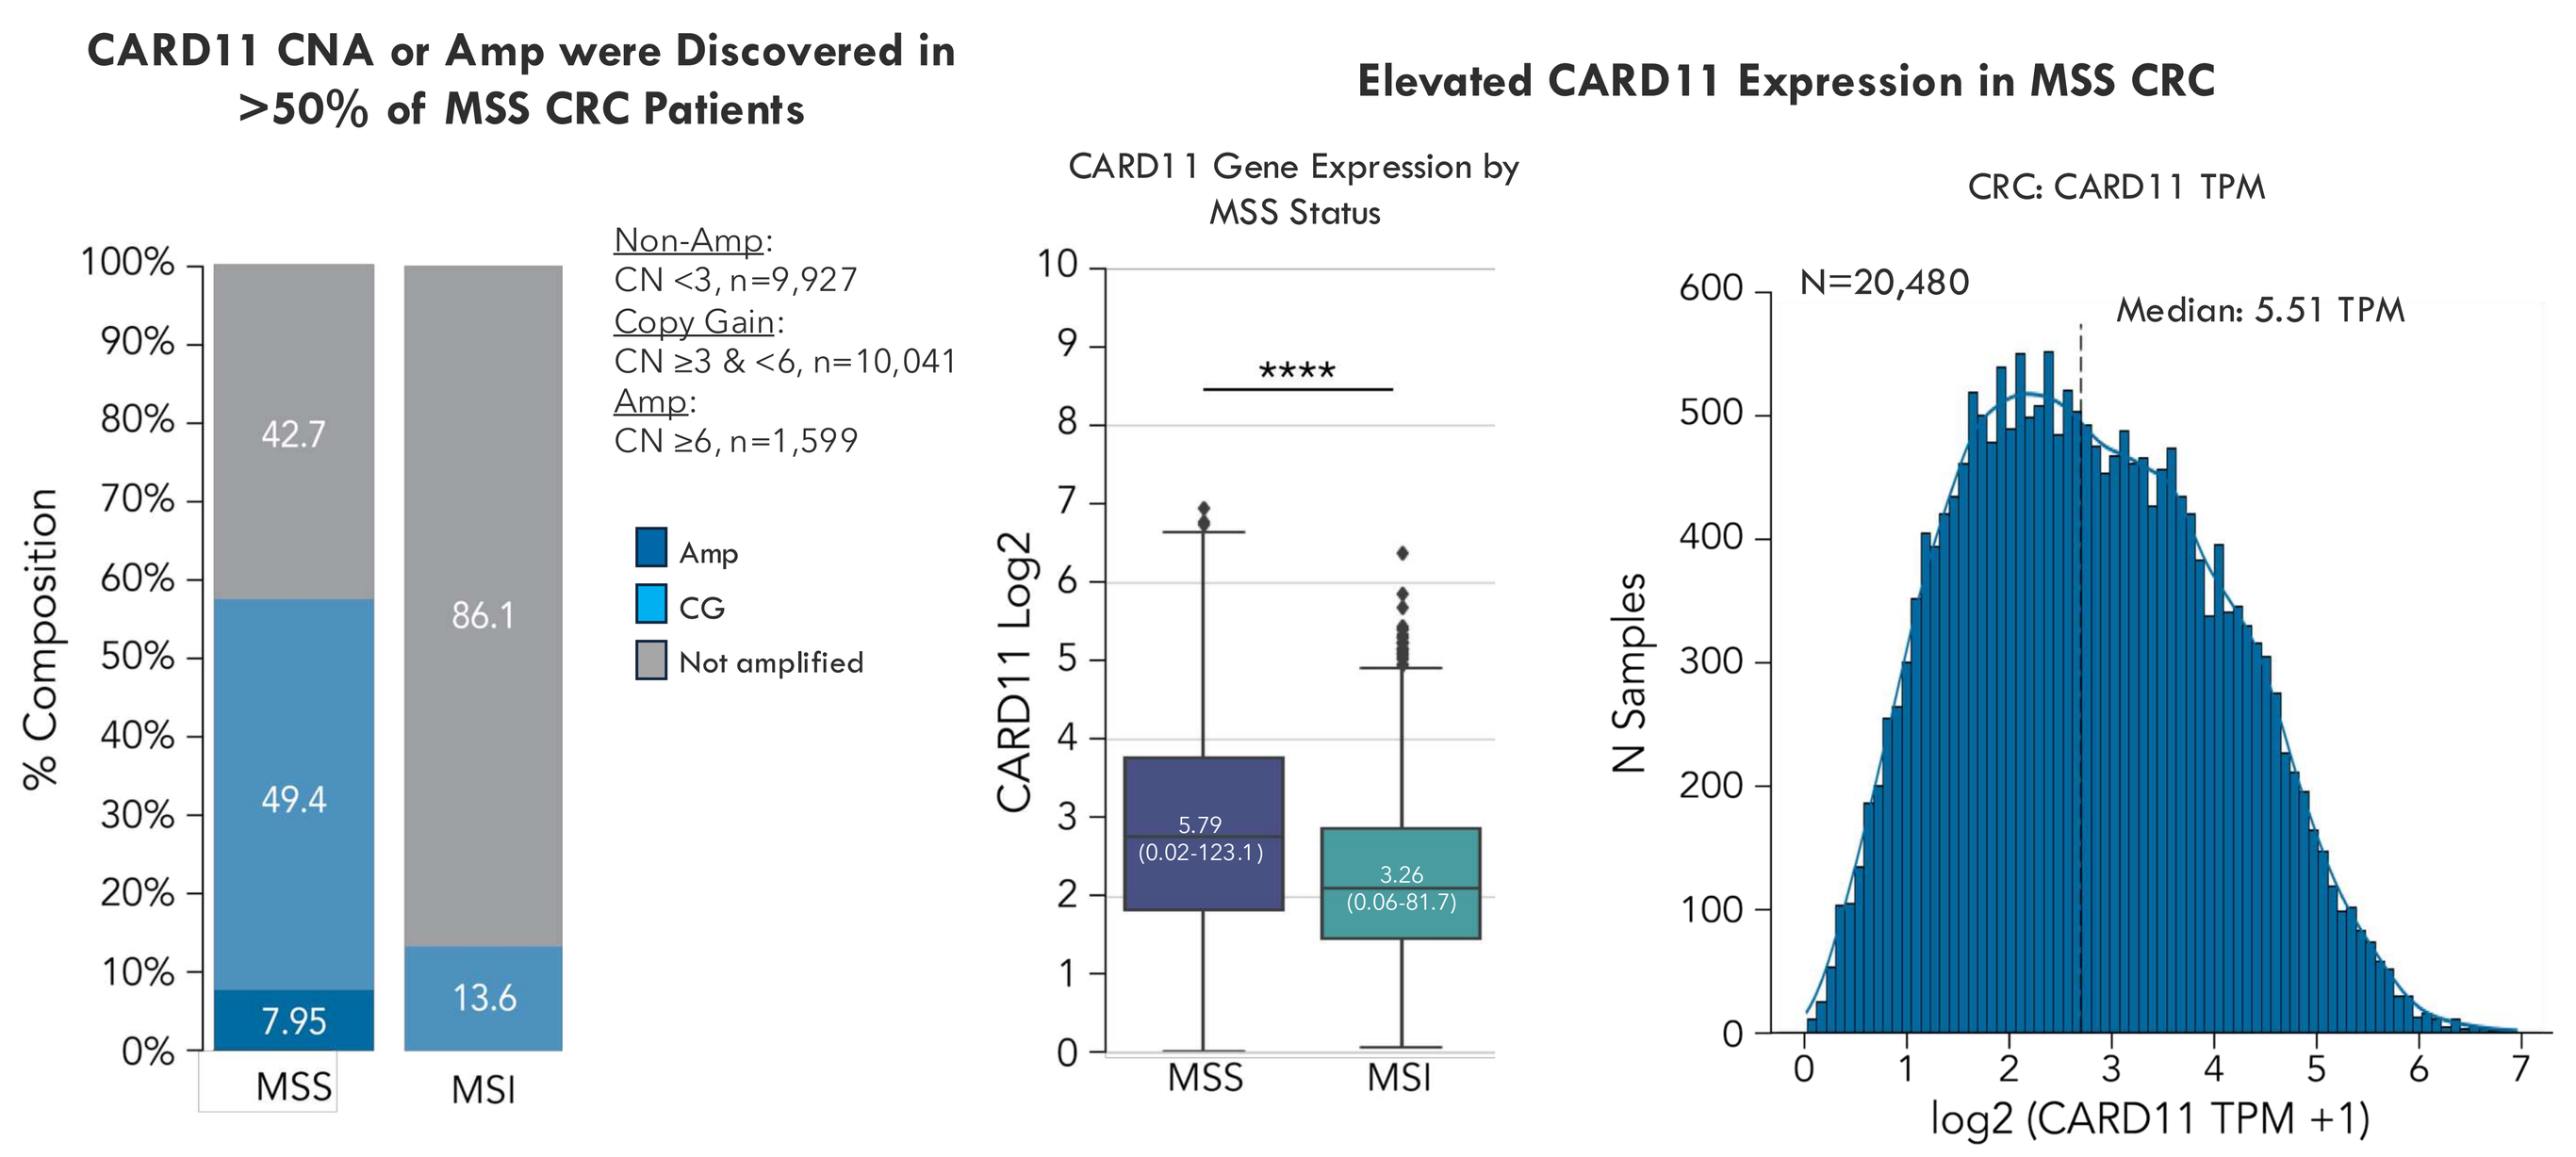
CARD11 CNA or Amp were Discovered in >50% of MSS CRC Patients
Non-Amp: CN <3, n=9,927
Copy Gain: CN ≥3 & <6, n=10,041
Amp: CN ≥6, n=1,599
Amp
CG
Not amplified
% Composition
42.7
49.4
7.95
86.1
13.6
MSS
MSI
Elevated CARD11 Expression in MSS CRC
CARD11 Gene Expression by MSS Status
CARD11 Log2

5.79 (0.02-123.1)
3.26 (0.06-81.7)
CRC: CARD11 TPM
N=20,480
Median: 5.51 TPM
N Samples
log2 (CARD11 TPM +1)

Same Database Revealed High CARD11 CRC has the Highest KRAS Activity Among Oncogenic Pathways (P< 0.0001)

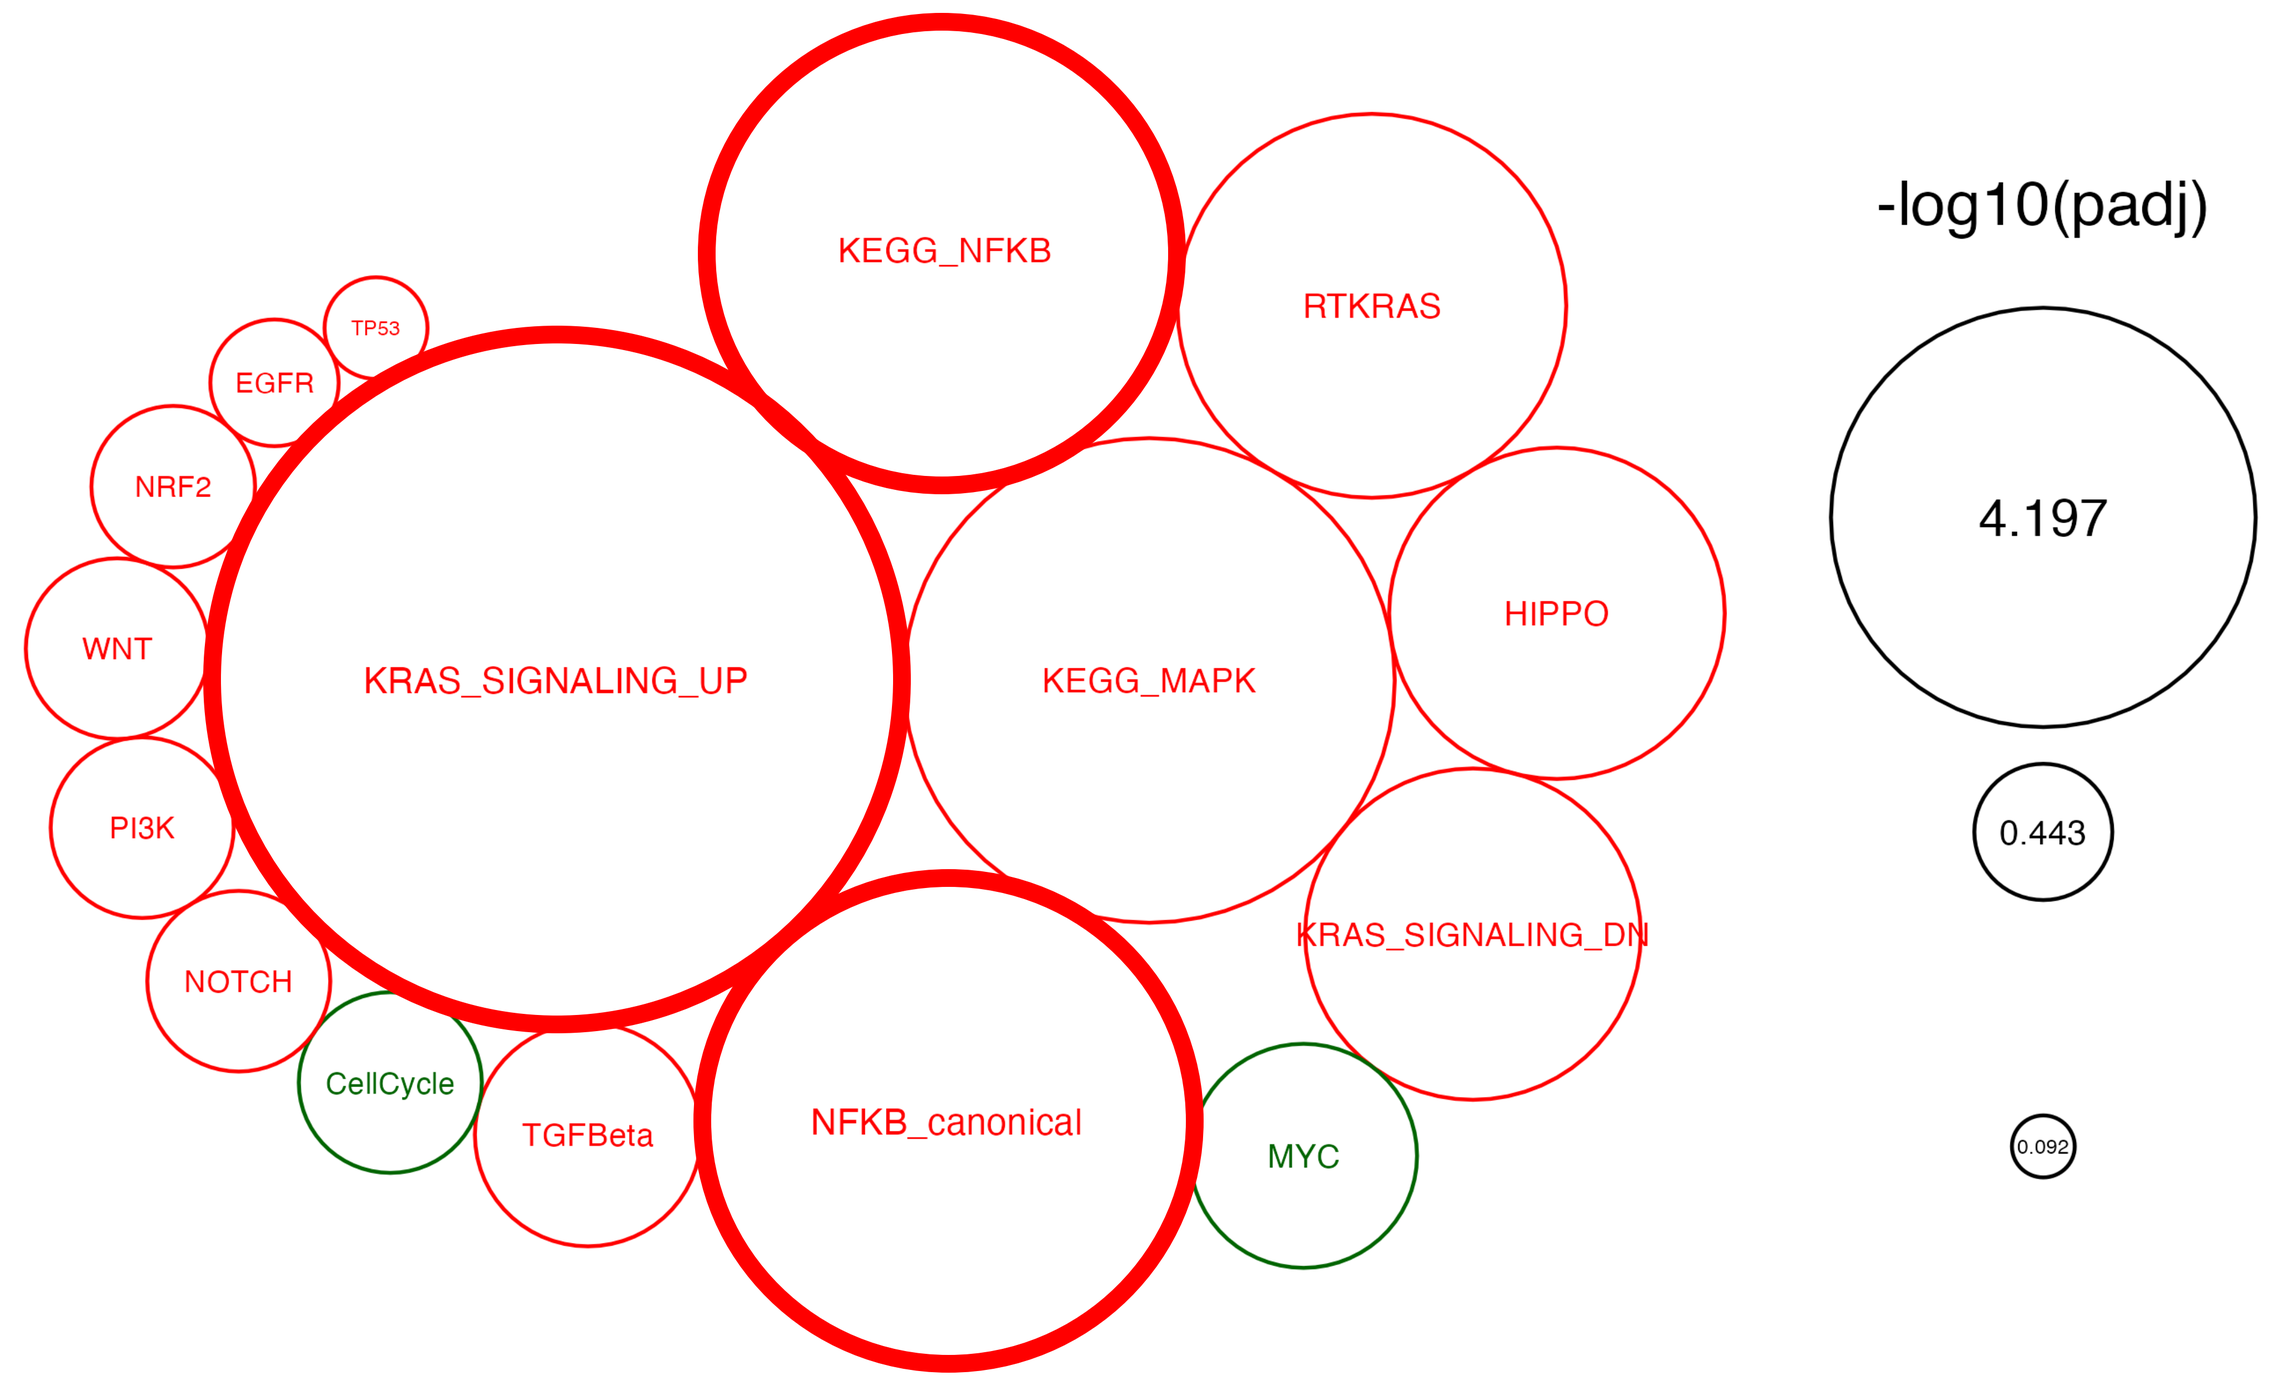
KEGG_NFKB
RTKRAS
TP53
EGFR
NRF2
WNT
KRAS_SIGNALING_UP
KEGG_MAPK
HIPPO
PI3K
KRAS_SIGNALING_DN
NOTCH
CellCycle
TGFBeta
NFKB_canonical
MYC
-log10(padj)
4.197
0.443
0.092

Top 25% expressing CARD11 CRC vs low 25% CARD11 expressing CRC.

CARD11 CRC had Shorter Time-on-Treatment of KRAS^{G12C} Inhibitor

Adagrasib/Sotorasib Time on Treatment (TOT)

- N=50 treated with Sotorasib (N=46 treated with only Sotorasib)
- N=35 treated with Adagrasib (N=31 treated with only Adagrasib)
- N=4 treated with both Sotorasib and Adagrasib

Statistics in Months	MSS	MSS, KRAS G12C	MSS, CARD11 Q1*	MSS, CARD11 Q4**
N#	62	53	15	14
Min	0	3	3	5
25%	18	26	24	21.5
Median	62	73	93	58.5
75%	124	134	136	88
Max	430	430	430	188

patients who received Sotorasib or Adgrasib in the group of interest
* Q1: Lowest 25% CARD11 expression
** Q4: Highest 25% CARD11 expression
^ Chi-square analyses on the table

Figure 2. CBM Inhibitor Selectively Induced Apoptosis in MSS/KRAS^{G12X} CRC

CBM Inhibitor Selectively Induced Apoptosis in MSS/KRAS^{G12X} CRC

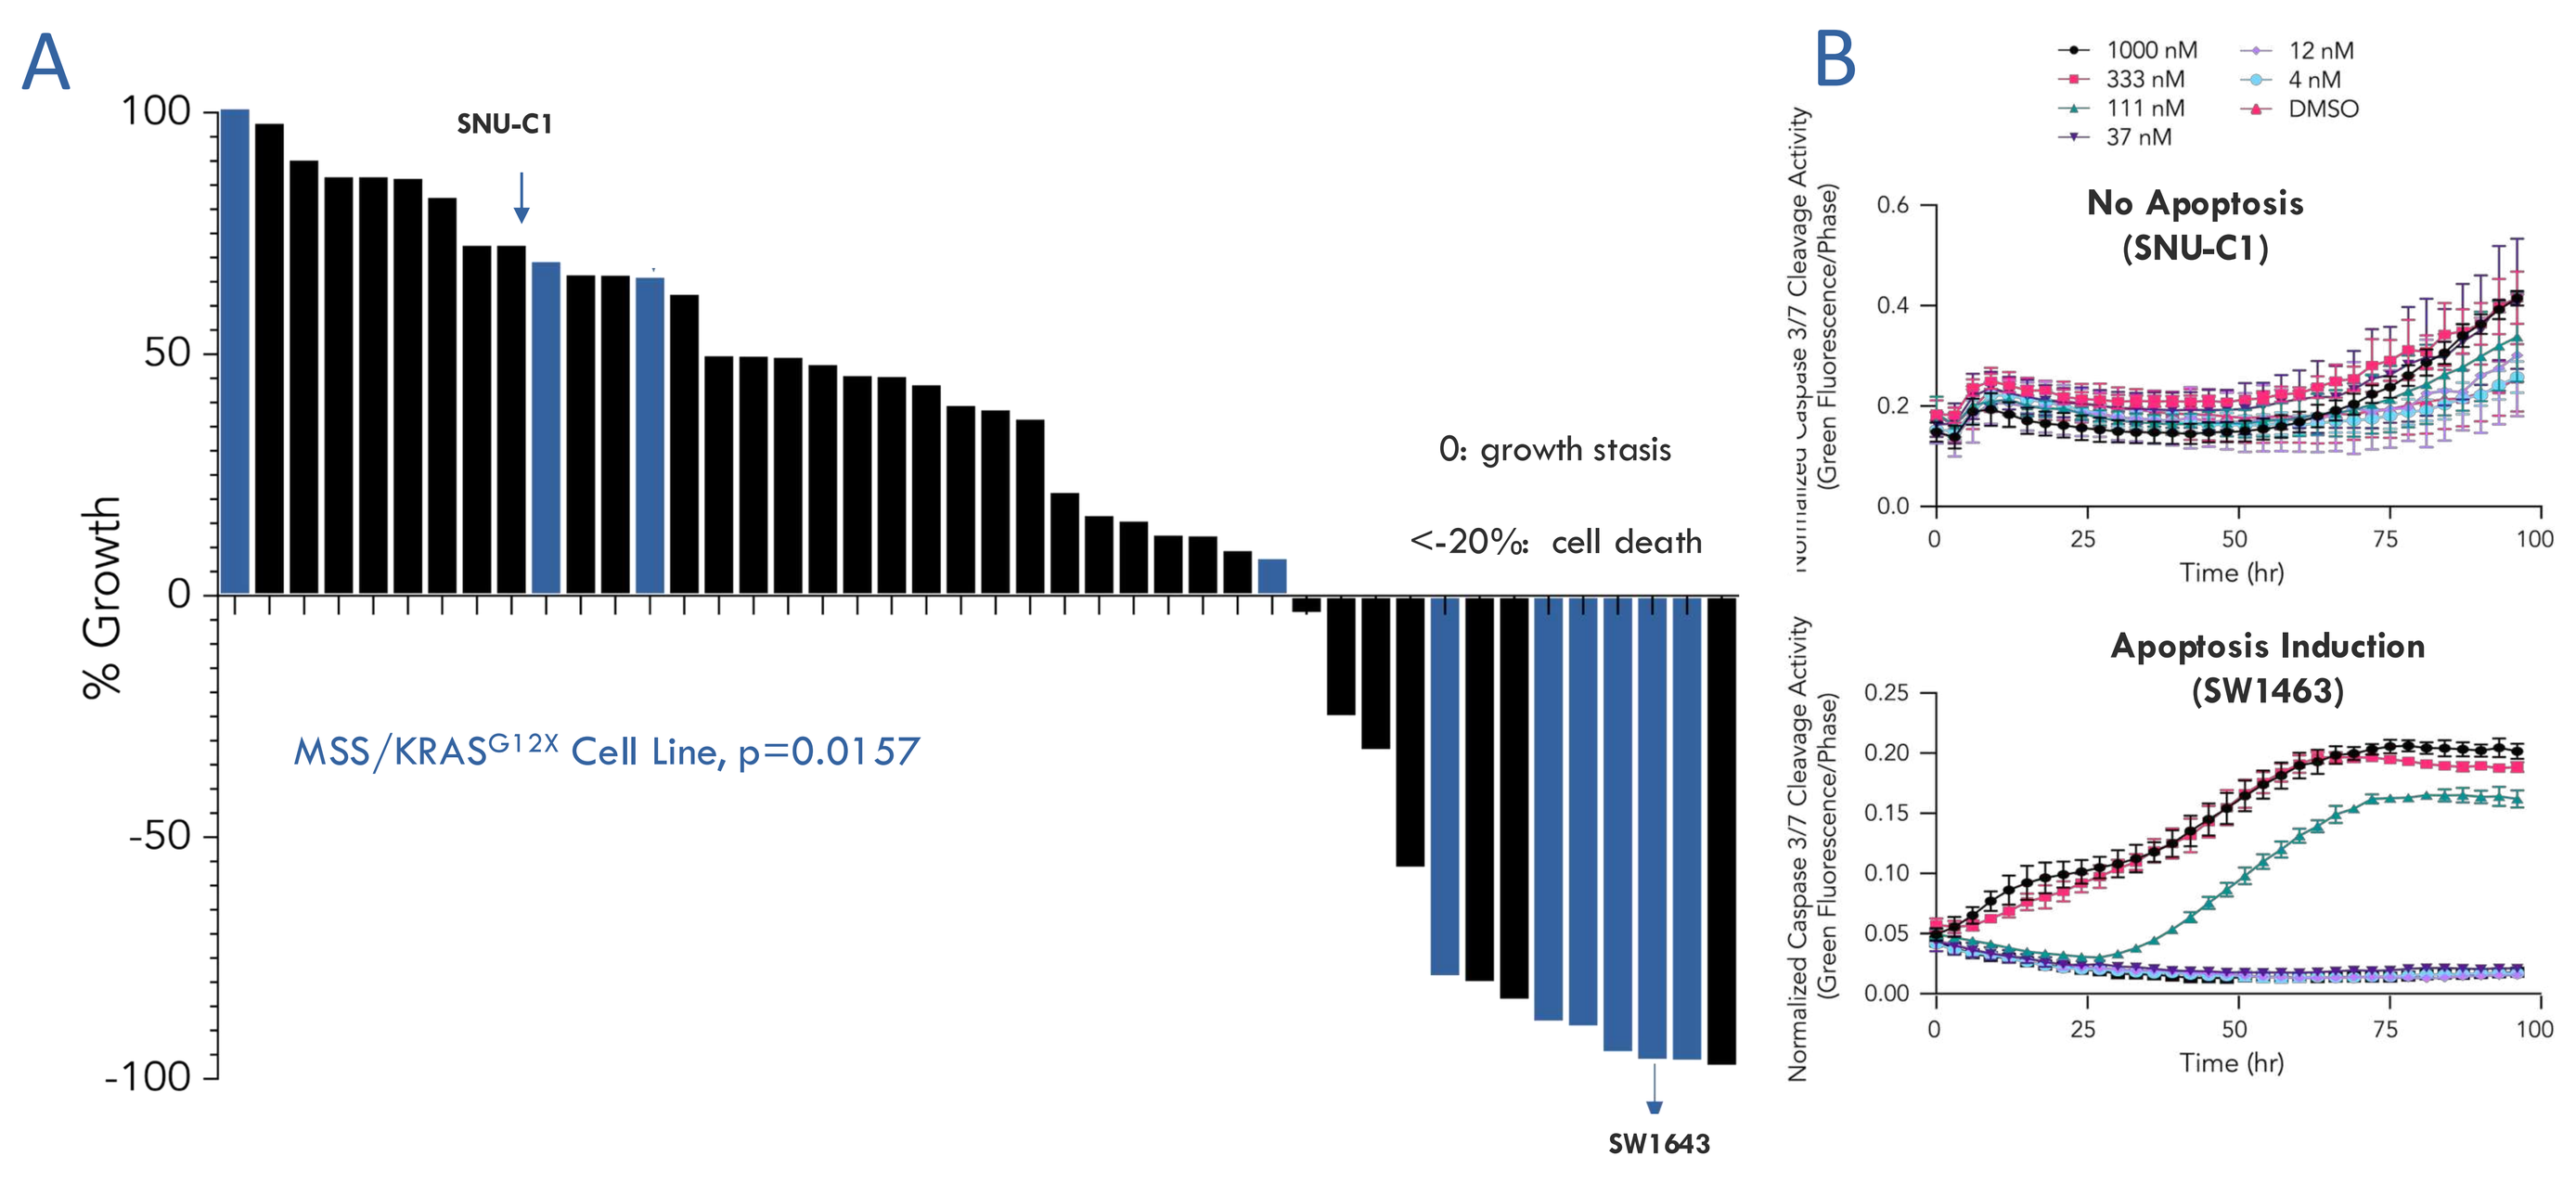
A
SNU-C1
% Growth
0: growth stasis
<-20%: cell death
MSS/KRASG12X Cell Line, p=0.0157
SW1643
B
1000 nM
333 nM
111 nM
37 nM
12 nM
4 nM
DMSO
No Apoptosis (SNU-C1)
Apoptosis Induction (SW1463)
Normalized Caspase 3/7 Cleavage Activity (Green Fluorescence/Phase)
Time (hr)

(A) A panel of CRC lines (including KRAS WT and KRAS mutant) were treated with CBM inhibitor (HOT-051) in dose and time-dependent manner. The plot was graphed based on 3 μM of HOT-051 (maximum inhibition) after 96hr treatment. 0-100: growth inhibition; 0: growth stasis; <-20%: cell death. (B) Representative cell lines for HOT-051 induced apoptosis. Dose and time-dependent caspase 3/7 cleavage was monitored using Incucyte.

Figure 3. CBM Inhibitor Induced Potent Apoptosis in MSS/KRAS^{G12X} CRC, Superior to KRASi Plus EGFR Antibody

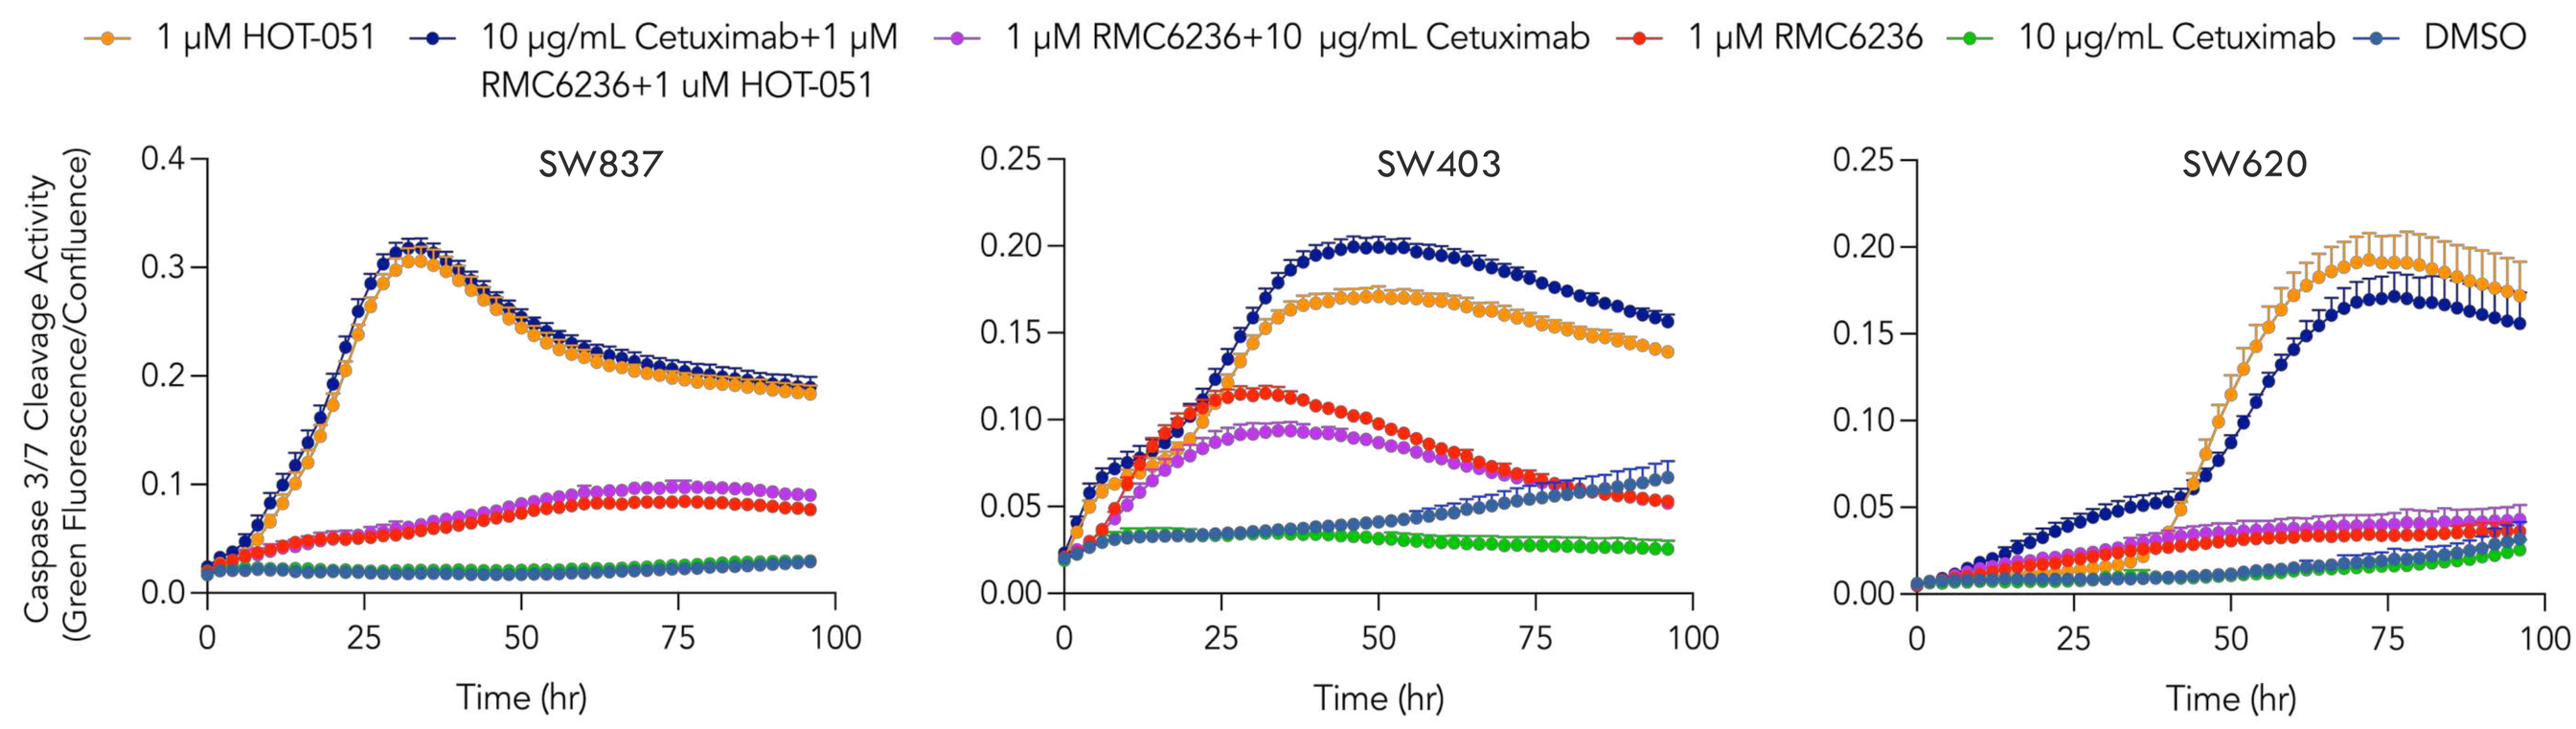
1 μM HOT-051
10 μg/mL Cetuximab+1 μM RMC6236+1 uM HOT-051
1 μM RMC6236+10 μg/mL Cetuximab
1 μM RMC6236
10 μg/mL Cetuximab
DMSO
SW837
SW403
SW620
Caspase 3/7 Cleavage Activity (Green Fluorescence/Confluence)
Time (hr)

Apoptosis was measured by caspase 3/7 cleavage using Incucyte.

Figure 4. CBM Inhibitors Blocked Multiple Oncogenic Survival Signals in Sensitive KRAS^{G12X} CRC

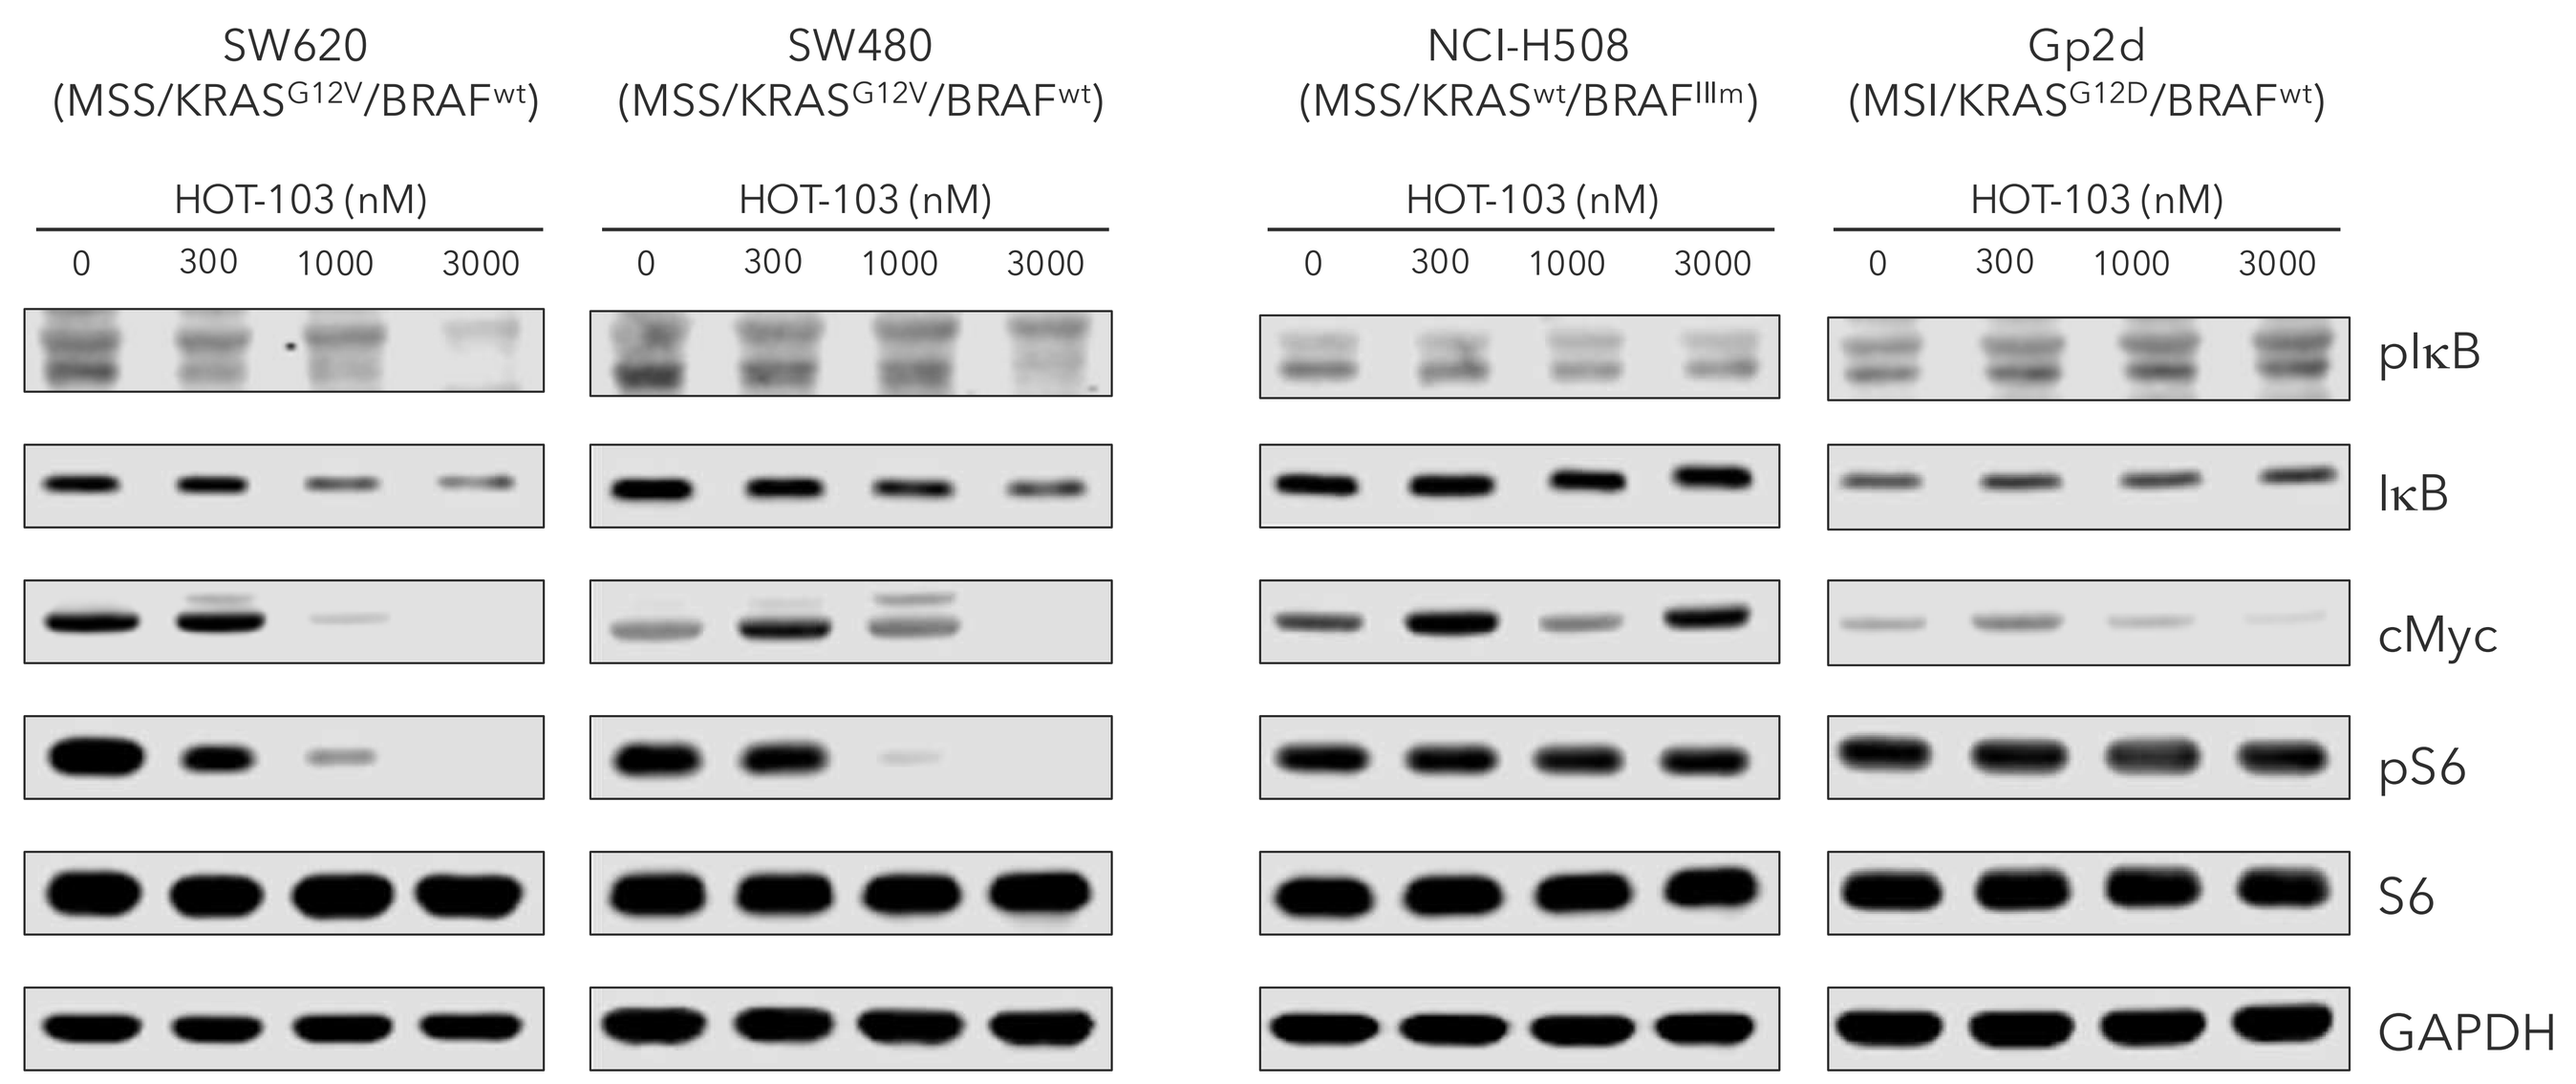
Sensitive CRC
Resistant CRC
SW620 (MSS/KRASG12V/BRAFwt)
SW480 (MSS/KRASG12V/BRAFwt)
NCI-H508 (MSS/KRASwt/BRAFIIIm)
Gp2d (MSI/KRASG12D/BRAFwt)
HOT-103 (nM)
0
300
1000
3000
pIκB
IκB
cMyc
pS6
S6
GAPDH

Cells were treated with HOT-103 (CBMi) for 24hr, followed by Western blot analysis of protein expression.

Figure 5. CBM Inhibitor Blocked Tumor Growth in Orthotopic SW620 (KRAS^{G12V}) CRC Model

Part 1: Tumor Regression After 7-day Treatment

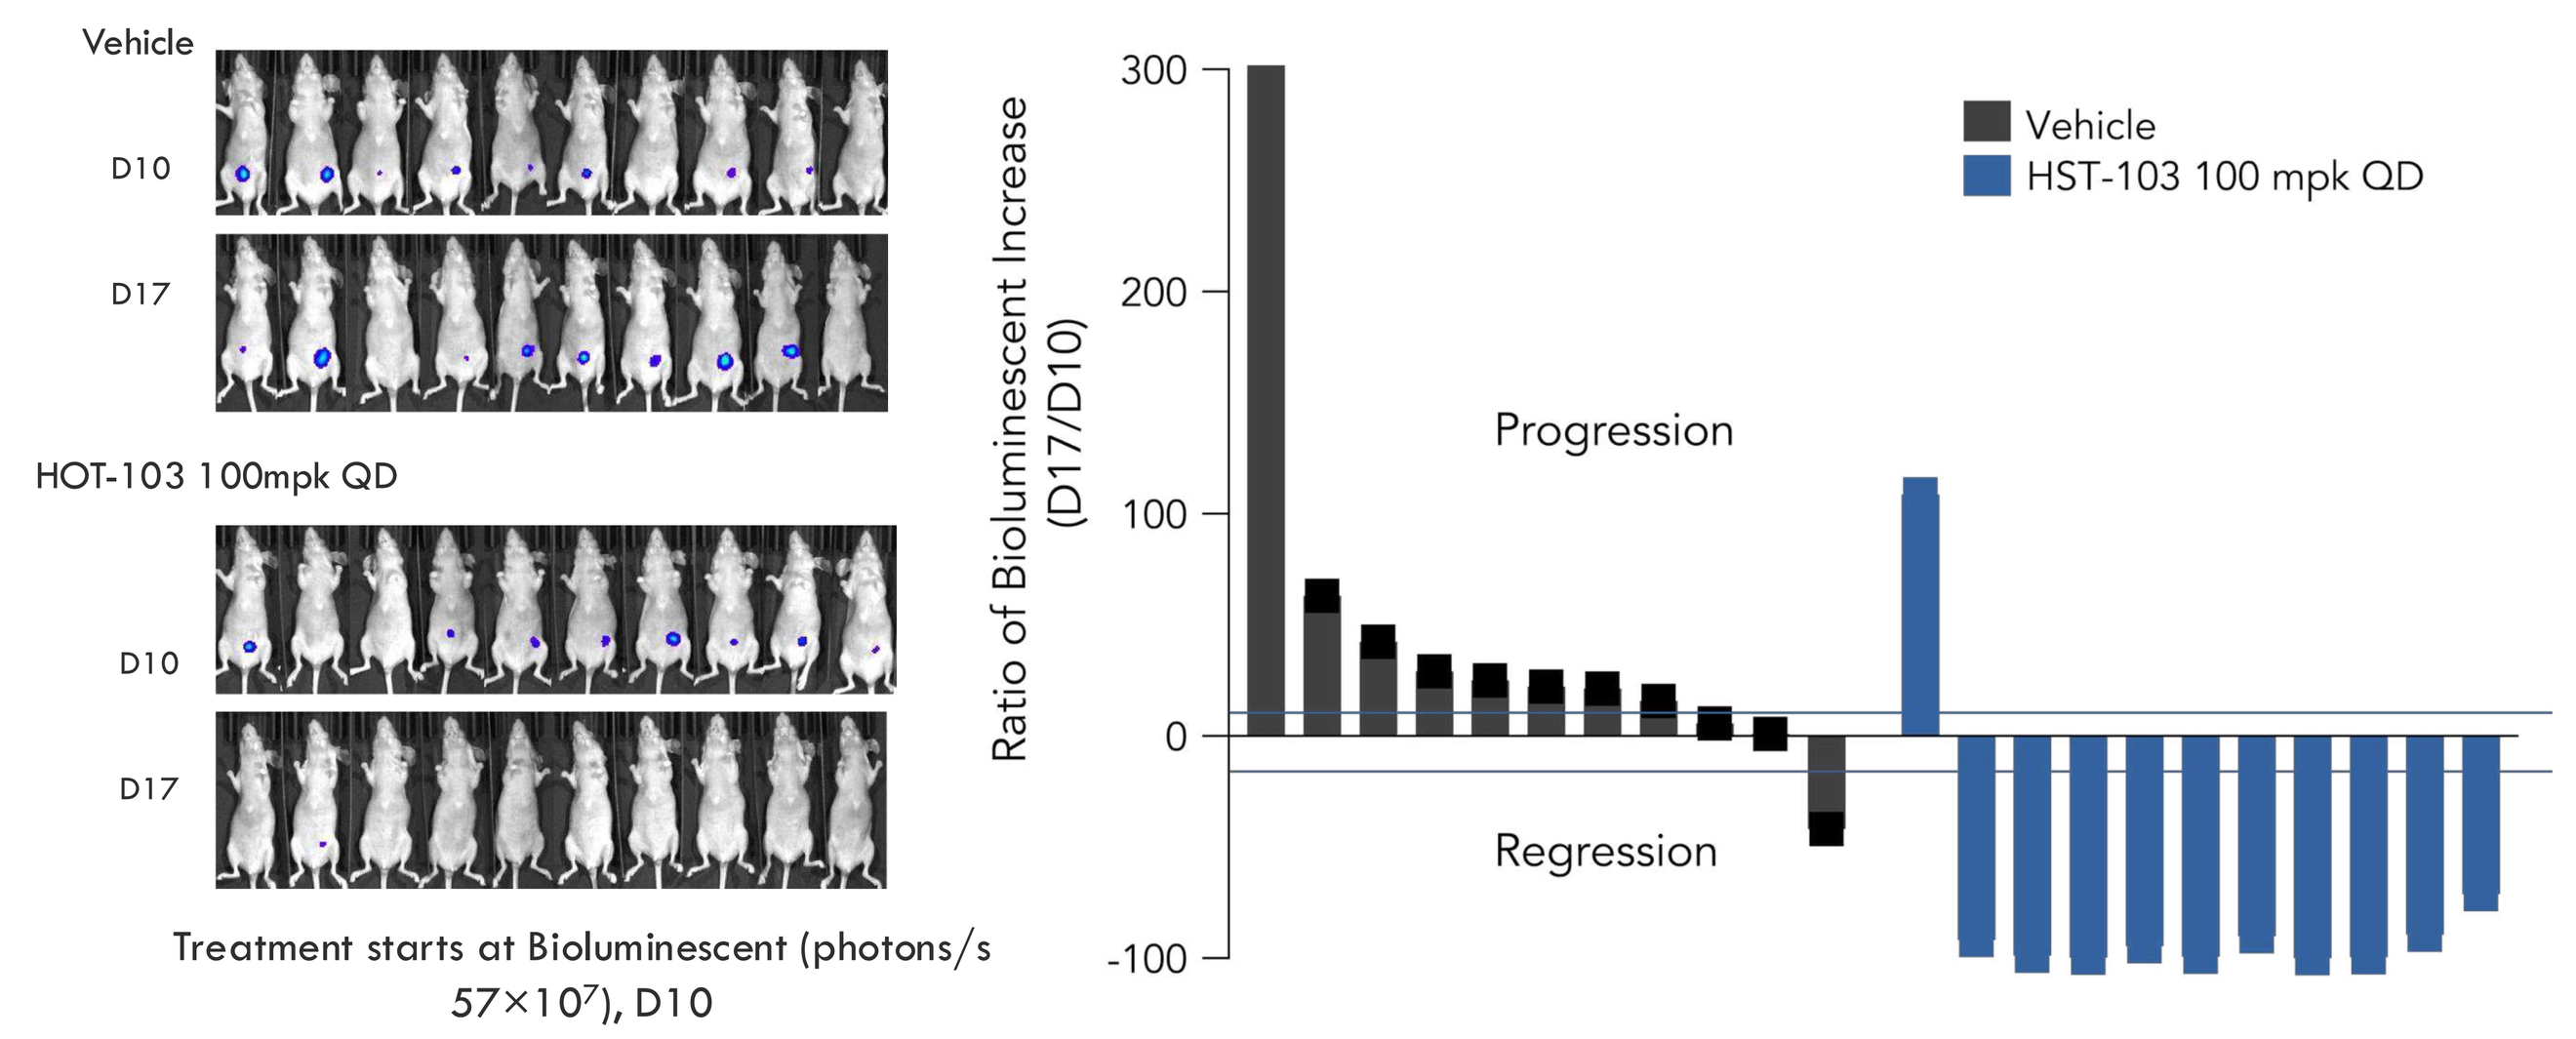
Vehicle
D10
D17
HOT-103 100mpk QD
Treatment starts at Bioluminescent (photons/s 57×10⁷), D10
Ratio of Bioluminescent Increase (D17/D10)
Vehicle
HST-103 100 mpk QD
Progression
Regression

Part 2: CBM Inhibitor Blocked Progression of High-Burden Tumors

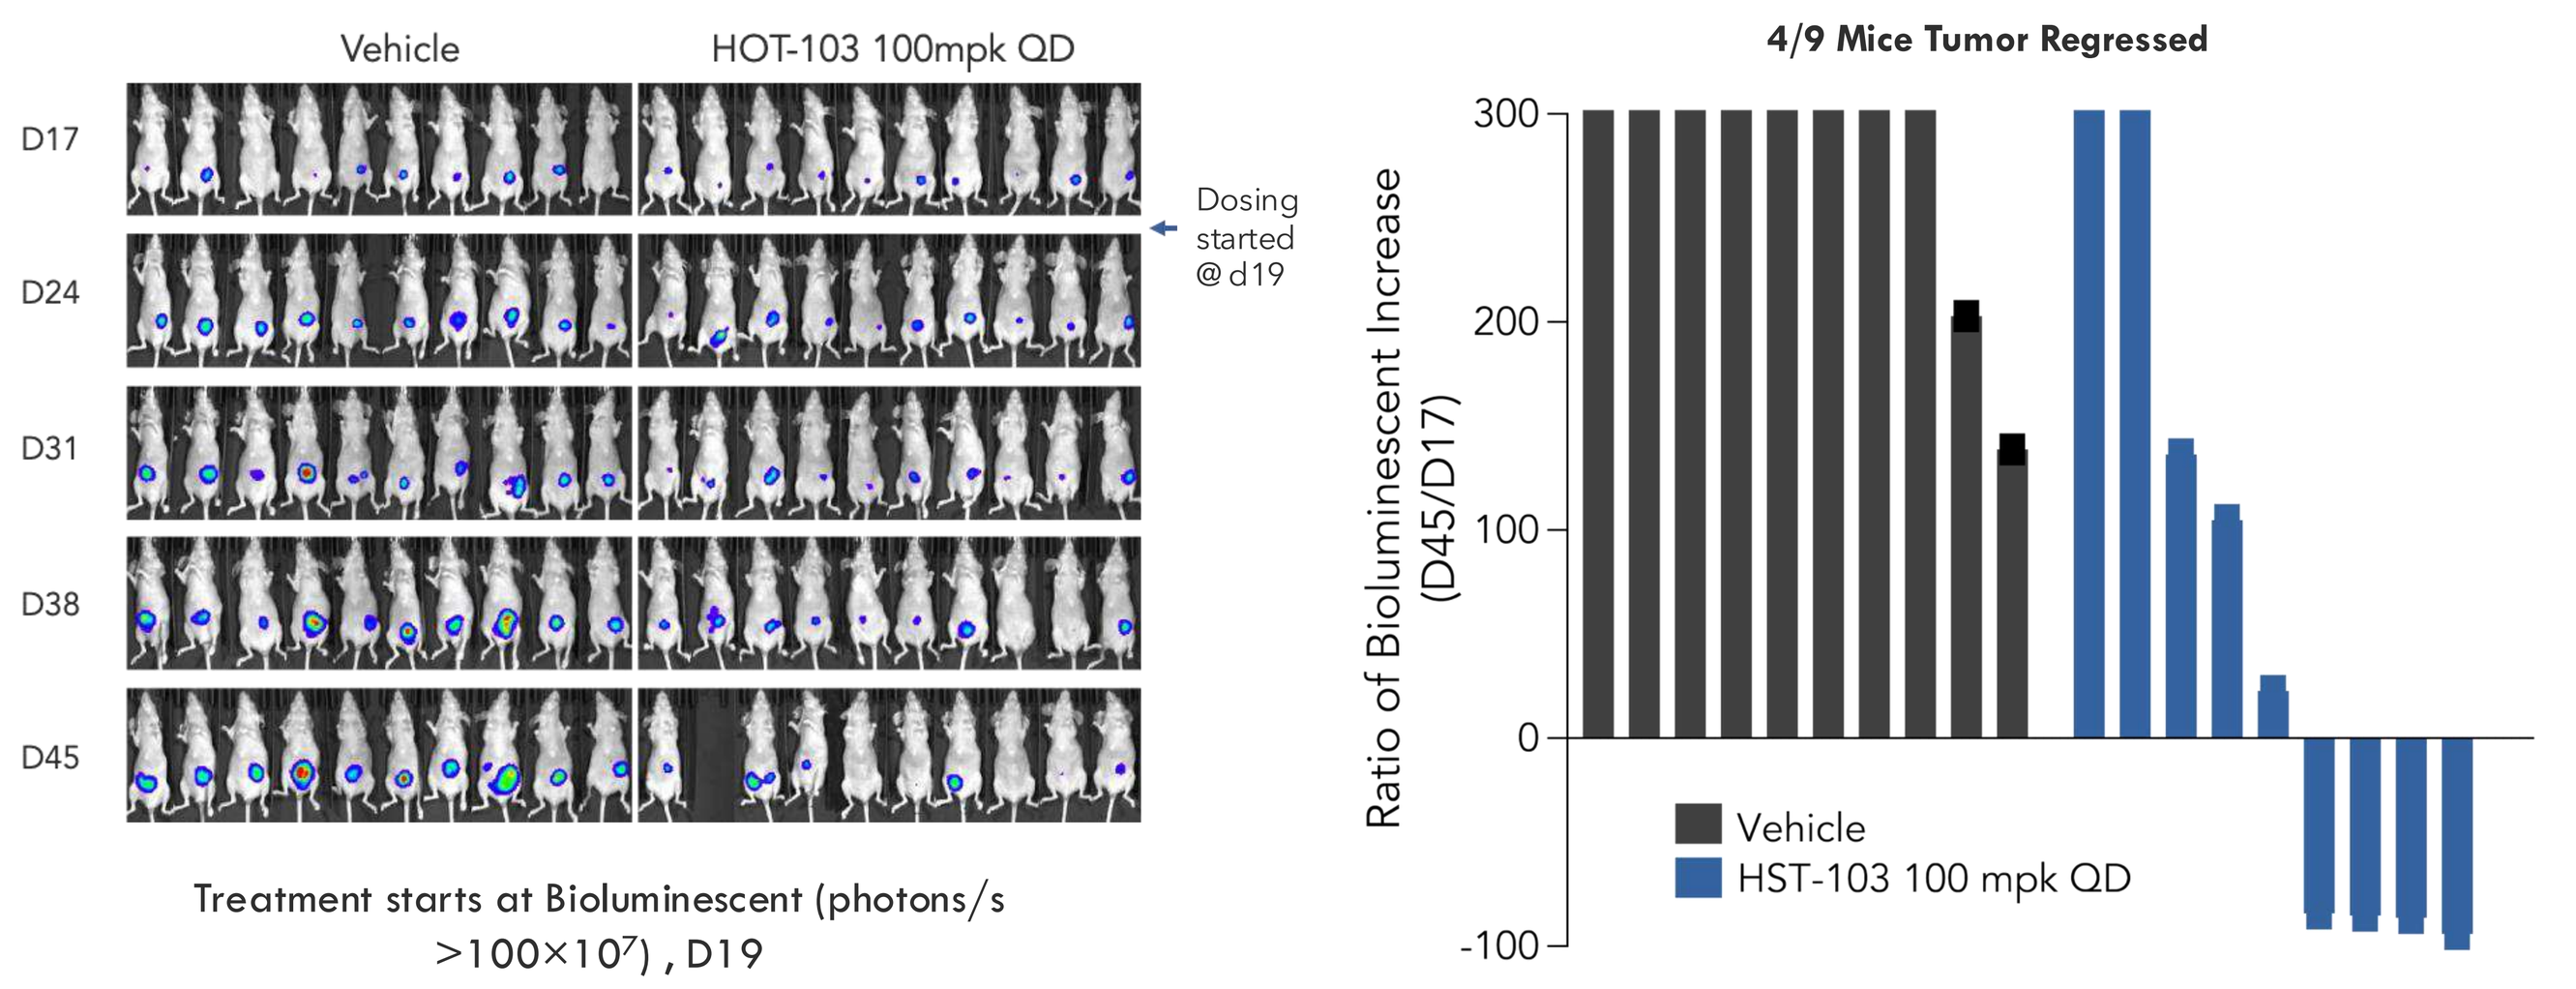
Vehicle
HOT-103 100mpk QD
D17
D24
D31
D38
D45
Dosing started @d19
Treatment starts at Bioluminescent (photons/s >100×10⁷), D19
4/9 Mice Tumor Regressed
Ratio of Bioluminescent Increase (D45/D17)
Vehicle
HST-103 100 mpk QD

Conclusions

- KRAS inhibitors do not induce strong apoptosis in vitro, in vivo, or in patients, especially for CRC, resulting in limited and non-durable response of KRASi.
- We discovered that KRAS^{G12X} CRC depends on the CBM signalosome for survival. Inhibition of CBM signalosome induced profound apoptosis/cell death of KRAS^{G12X} CRC in *in vitro* and *in vivo* preclinical models.
- CBMi plus KRASi may provide deep and durable responses by targeting two cancer hallmarks: proliferation & survival.

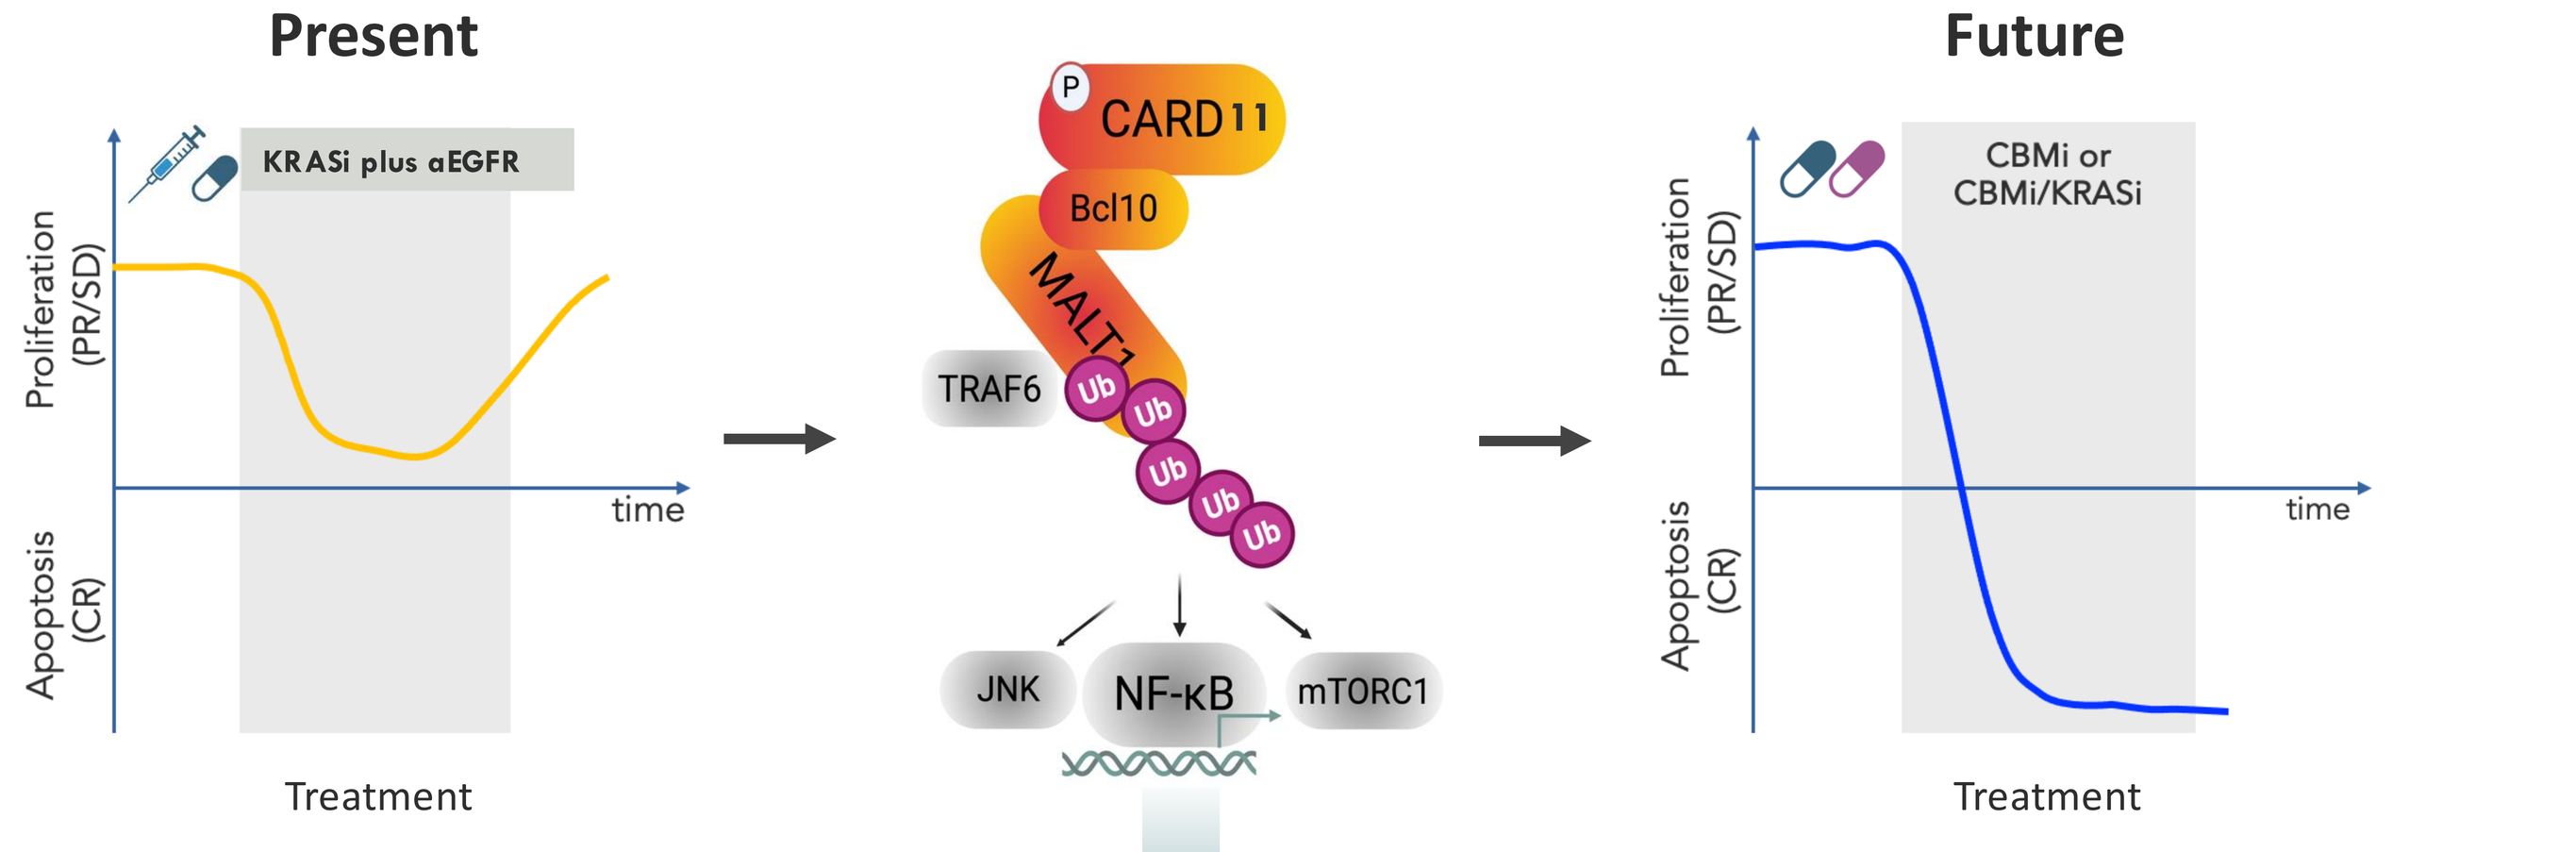
Present
KRASi plus αEGFR
Proliferation (PR/SD)
Apoptosis (CR)
time
Treatment
CARD11
Bcl10
MALT1
TRAF6
JNK
NF-κB
mTORC1
Future
CBMi or CBMi/KRASi
Treatment

Complete inhibition of MAPK to have strong anti-proliferation effect

Pro-survival signals for KRAS^{G12X} CRC

- Complete inhibition of MAPK and multiple oncogenic signals (NFκB, mTORC1 and C-MYC)
- Anti-proliferation+ apoptosis effect for deep & durable responses

HotSpot Therapeutics, Inc • One Design Center Place • Suite 19-600 • Boston, MA 02210, USA • hotspotthera.com
Contact: Natalie Wildenradt - nwildenradt@hotspotthera.com
ESMO Gastrointestinal Cancers Congress • Barcelona, Spain • July 2-5, 2025